Pharmacokinetic and Pharmacodynamic Assessments with ITCA 650, Continuous Subcutaneous Delivery of Exenatide, in Type 2 Diabetes

J. Dahms[1], Y. Chandrasekher[1], R. Fielding[2], R. Zhou[3], R. Henry[4], J. Rosenstock[5], T. Alessi[1], K. Luskey[1]

Abstract

Background and Aim: ITCA 650 is a subcutaneous osmotic mini-pump that provides continuous delivery of exenatide. ITCA 650 has been shown to reduce FPG, HbA1c and body weight (BW). The relationship between the PK and PD of ITCA 650 was examined.

Materials and methods: PK (plasma exenatide concentrations) and PD (FPG, HbA1c, BW) data were collected from type 2 diabetics in a 28-day Phase 1b study (44 subjects) and a 24-week Phase 2 study (155 metformin-treated subjects) with a 24-week extension period. ITCA 650 doses ranged from 10-80 mcg/day in Phase 1b and 20-80 mcg/day in Phase 2. Css and AUCss were assessed from 24 hrs after placement through end of treatment. Exposure-response relationships were assessed with linear and tobit regression models, and graphically explored with LOESS (locally weighted scatterplot smoothing) techniques.

Results: There was a dose-proportional increase in exenatide exposure (AUCss and Css). Mean Css ranged from 38 ± 12 pg/mL at 10 mcg/day to 305 ± 199 pg/mL at 80 mcg/day. ITCA 650 provided continuous exposure for the duration of each placement (up to 12 weeks) and for the duration of treatment (up to 48 weeks). Exposures with ITCA 650 were consistent with exposures reported for SC infusion and BID exenatide injection. In contrast to BID exenatide injections, a pattern of peak and trough levels was not observed with ITCA 650. The effects of ITCA 650 were apparent within 24 hrs of placement. Plasma exenatide was detected in 32 of 44 Phase 1 subjects within 6-12 hrs, and steady state concentrations were achieved within 24-48 hrs in all dose groups. Reductions in FPG were observed within one day. FPG and 2-hr postprandial glucose were significantly decreased in the 20, 40 and 80 mcg/day dose groups within 1-5 days of initiating ITCA 650 and throughout the 28-day treatment period ($p<0.05$).
The PK/PD relationship of exenatide exposure relative to changes in HbA1c and BW was examined in Phase 2. All doses (20-80 mcg/day) were active suggesting an ED_{50} of < 20 mcg/day. Higher exenatide concentrations tended to produce larger reductions in HbA1c and BW; concentrations associated with all doses were on the relatively flat upper part of the response curve. PD responses were maintained over 48 weeks of treatment.
Baseline HbA1c also contributed to the change in HbA1c. In the combined population from all dose groups, subjects in the lowest quartile of starting HbA1c values had a mean HbA1c of 7.0% and a change in HbA1c of -0.7% whereas subjects in the highest quartile had a mean HbA1c of 9.2% and a change in HbA1c of -1.9%.
Consistent with its 2.4 hr half-life, exenatide was not measurable in any subject 24 hrs after ITCA 650 removal. Samples collected 7 days after ITCA 650 removal confirmed that FPG levels had returned to baseline values.

Conclusion: ITCA 650 provided continuous exposure for up to 48 weeks, dose-proportional exposures, rapid attainment of steady state plasma exenatide following placement and rapid clearance of exenatide after removal. A relationship between exenatide exposure and glycemic and weight parameters was noted. In addition, the baseline HBA1c played a role in the magnitude of the response to ITCA 650 treatment.

Background and Aim

- ITCA 650 is a subcutaneous osmotic mini-pump that provides continuous zero-order delivery of exenatide
- Dosing for up to 12 months with a single placement; 3-month ITCA 650s used for these studies
- Placement/removal in a 10-minute in-office procedure
- Aim: To evaluate PK / PD with ITCA 650

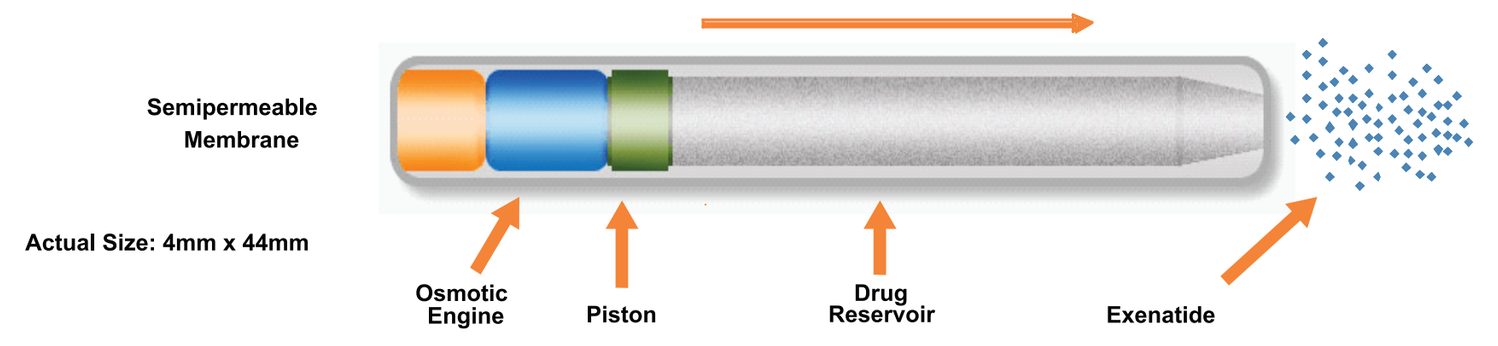
Semipermeable Membrane
Actual Size: 4mm x 44mm
Osmotic Engine
Piston
Drug Reservoir
Exenatide

Exenatide Release from ITCA 650 at 37°C after 12-Month Storage at 25°C

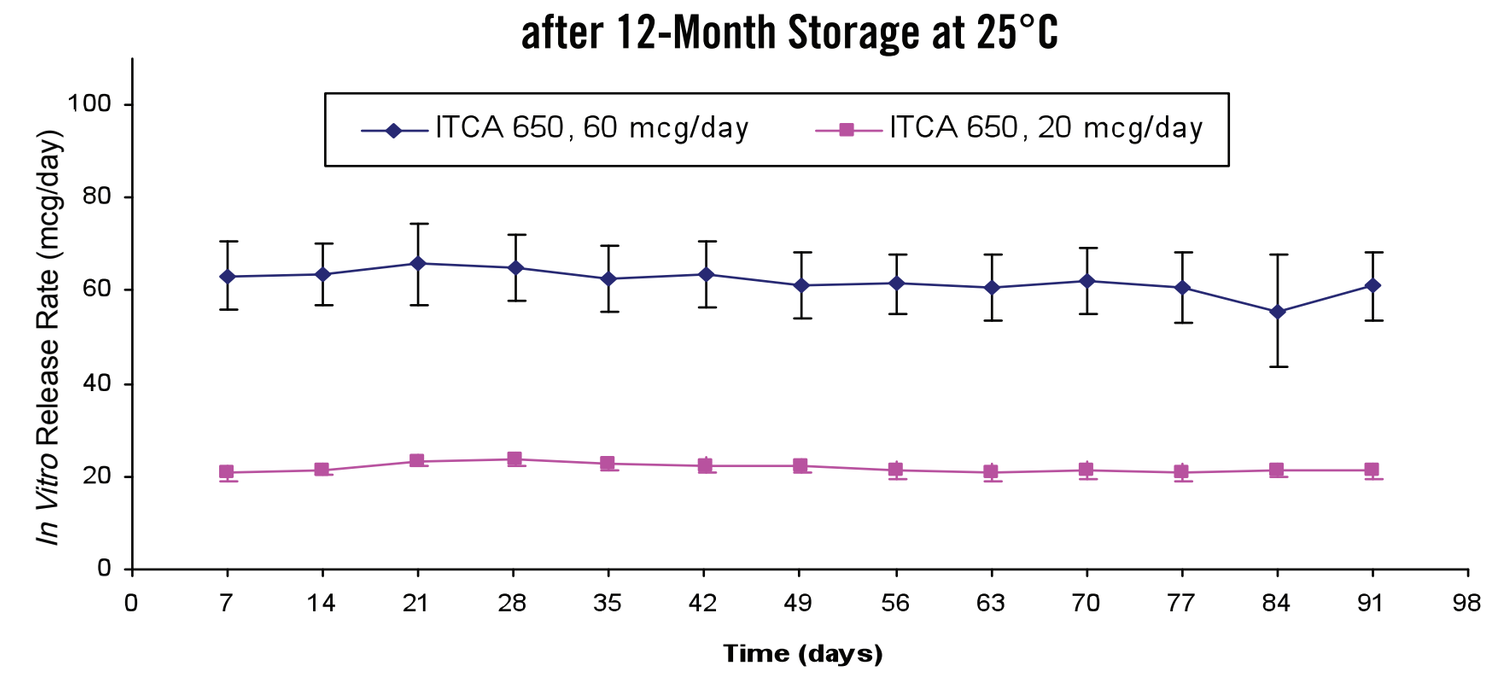
ITCA 650, 60 mcg/day
ITCA 650, 20 mcg/day
In Vitro Release Rate (mcg/day)
Time (days)

Methods

- PK and PD data were collected from type 2 diabetics in a 28-day Phase 1b study (44 pts) and a 48-week Phase 2 study (155 metformin-treated pts). ITCA 650 doses ranged from 10-80 mcg/day in Phase 1b and 20-80 mcg/day in Phase 2
- C_{SS} and AUC_{SS} were assessed from 24 hrs after placement through end of treatment. Plasma exenatide concentrations were determined using a validated ECL assay with range of quantitation from 29.3 pg/mL to 15,000 pg/mL
- Exposure-response relationships were assessed with linear and tobit regression models, and graphically explored with LOESS (locally weighted scatterplot smoothing) techniques

Pharmacokinetics with ITCA 650

- Exposures are dose-proportional

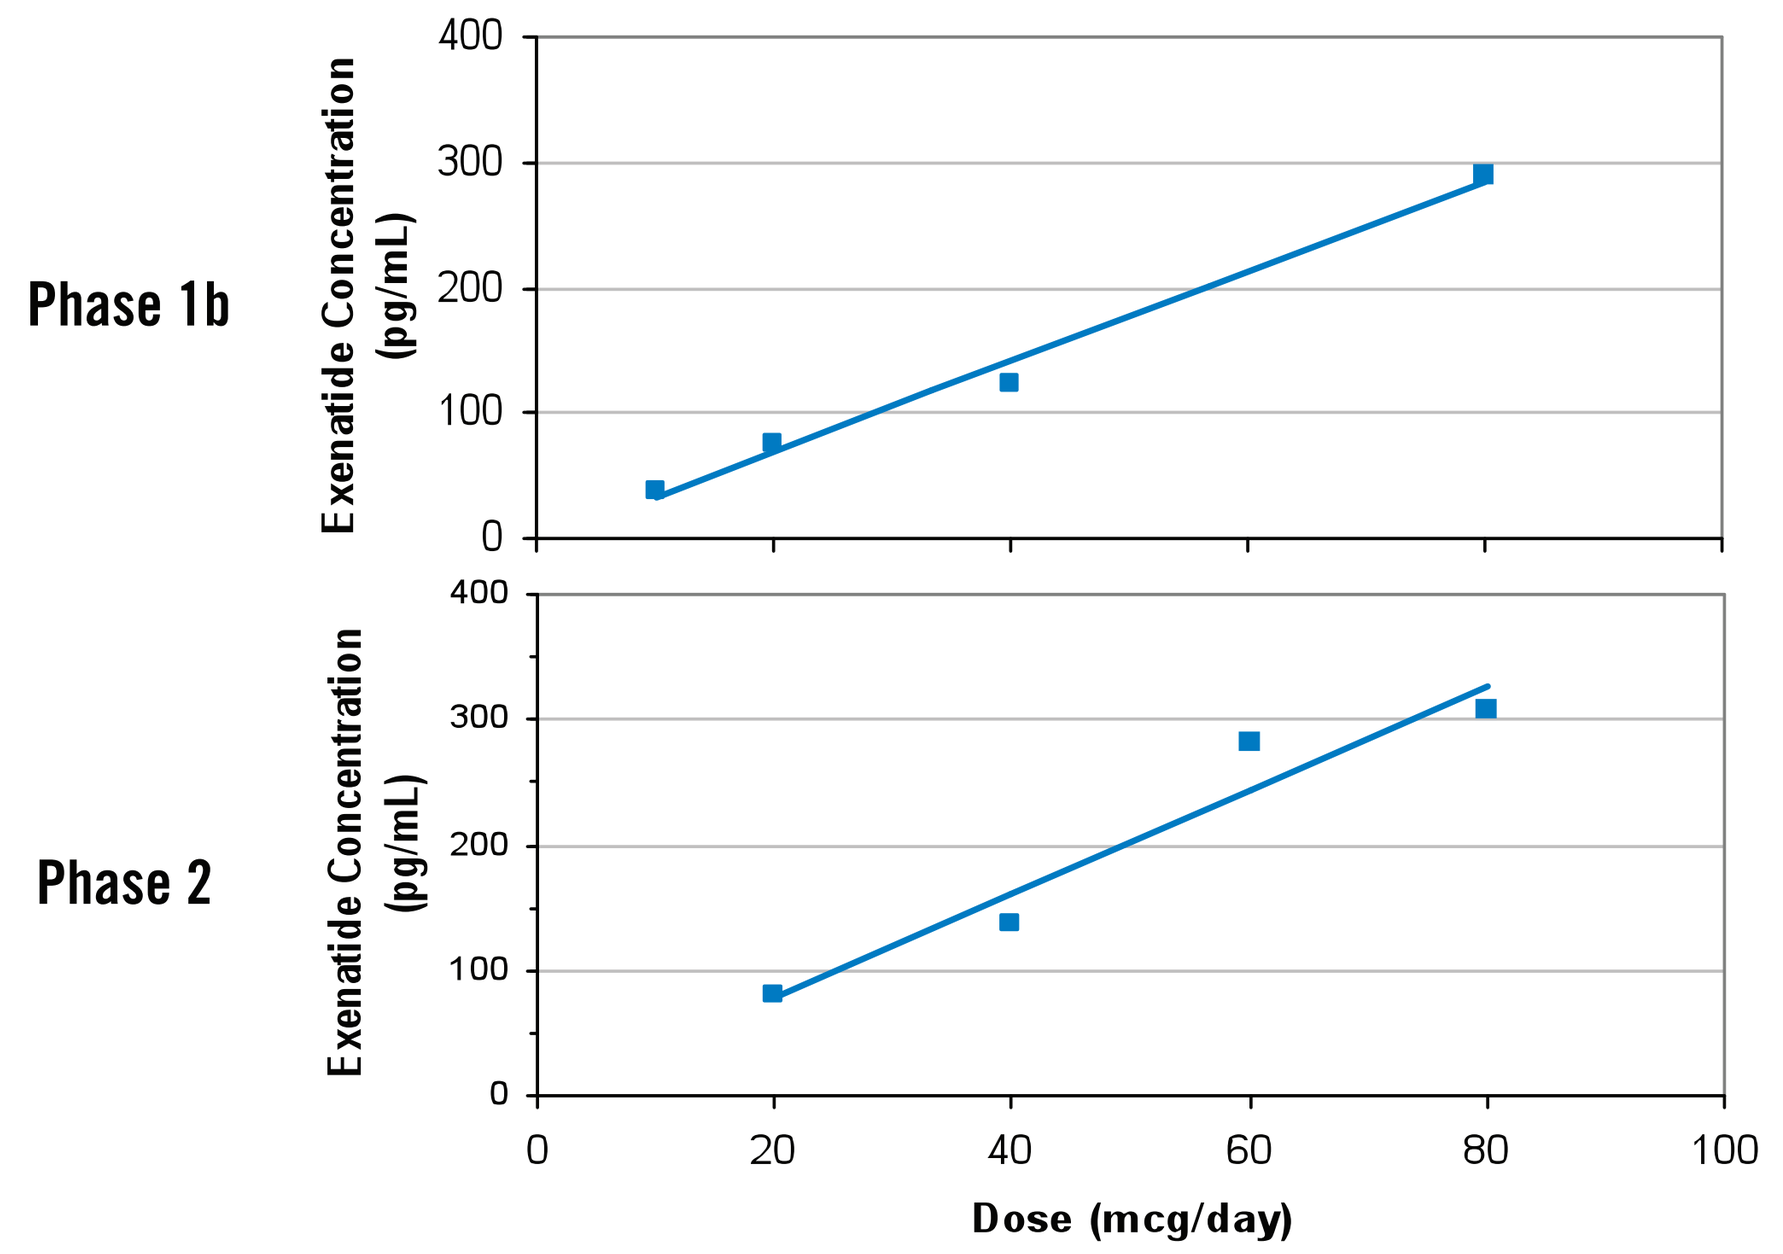
Phase 1b
Phase 2
Exenatide Concentration (pg/mL)
Dose (mcg/day)

C_{ave} based on the average of each subject's mean concentration from 24 hours after ITCA 650 placement until removal.

Comparison to Infusion, Injection

- ITCA 650 produced plasma exenatide levels and standard deviations similar to sc infusion
- Bioavailability of exenatide by ITCA 650 was similar to that observed with immediate release exenatide injection at the same dose
- Steady state concentrations with ITCA 650 at 60 mcg/day were similar to Cmax reported with immediate release exenatide 10 mcg injection

Parameter	ITCA 650 60 mcg/day	Exenatide Infusion[1] 60.6 mcg/day
C_{ave} (mean (SD)) (pg/mL)	278.2 (155.2)	270.1 (118.4)

Parameter Mean (SD)	ITCA 650 20 mcg/day	ITCA 650 60 mcg/day	Exenatide Injection[2] 10 mcg bolus	Exenatide Injection[2] 20 mcg bolus
C_{max} (pg/mL)	--	--	241	342
C_{ave} (pg/mL)	81.1	278	--	--
AUC (pg·hr/mL)[3]	1946	6672	1046	1963

1 Taylor K, et al. Day-long subcutaneous infusion of exenatide lowers glycemia in patients with type 2 diabetes. Horm Metab Res. 2005 Oct;37(10):627-32
2 Kolterman, OG, et al. Pharmacokinetics, pharmacodynamics, and safety of exenatide in patients with type 2 diabetes mellitus. Am J Health-Syst Pharm. 2005 Jan; 62: 173-181
3 AUC_{ave} for ITCA 650, $AUC_{(0\text{-}15\ hr)}$ for Byetta

Pharmacodynamics of ITCA 650

- HbA1c at Baseline and Weeks 24 and 48

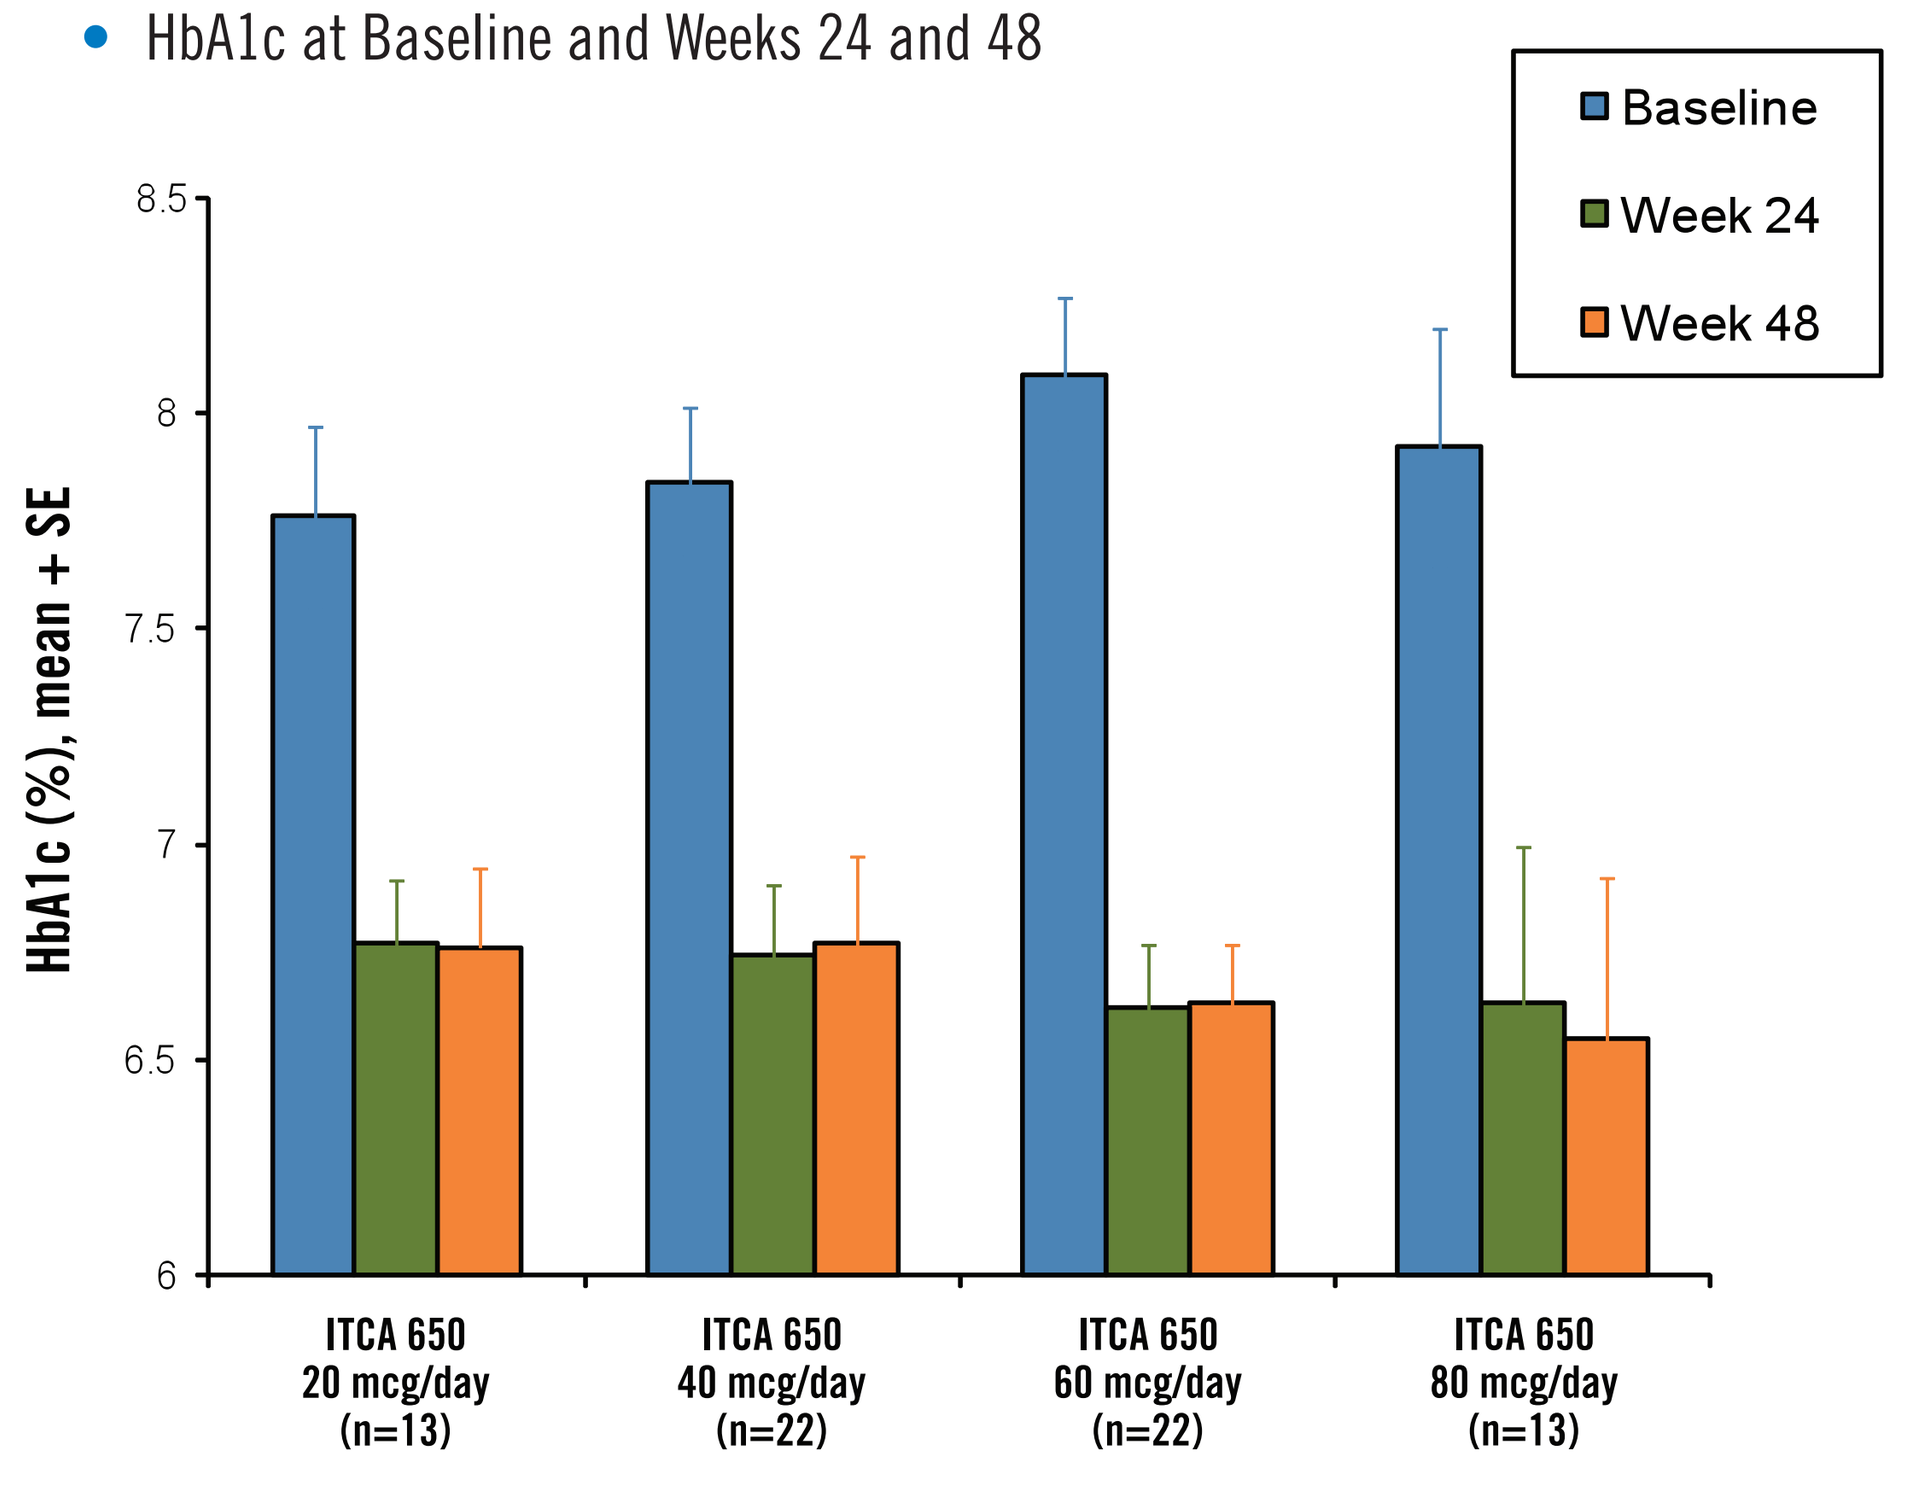
Baseline
Week 24
Week 48
HbA1c (%), mean + SE
ITCA 650 20 mcg/day (n=13)
ITCA 650 40 mcg/day (n=22)
ITCA 650 60 mcg/day (n=22)
ITCA 650 80 mcg/day (n=13)

- Higher baseline HbA1c is associated with a greater reduction in HbA1c

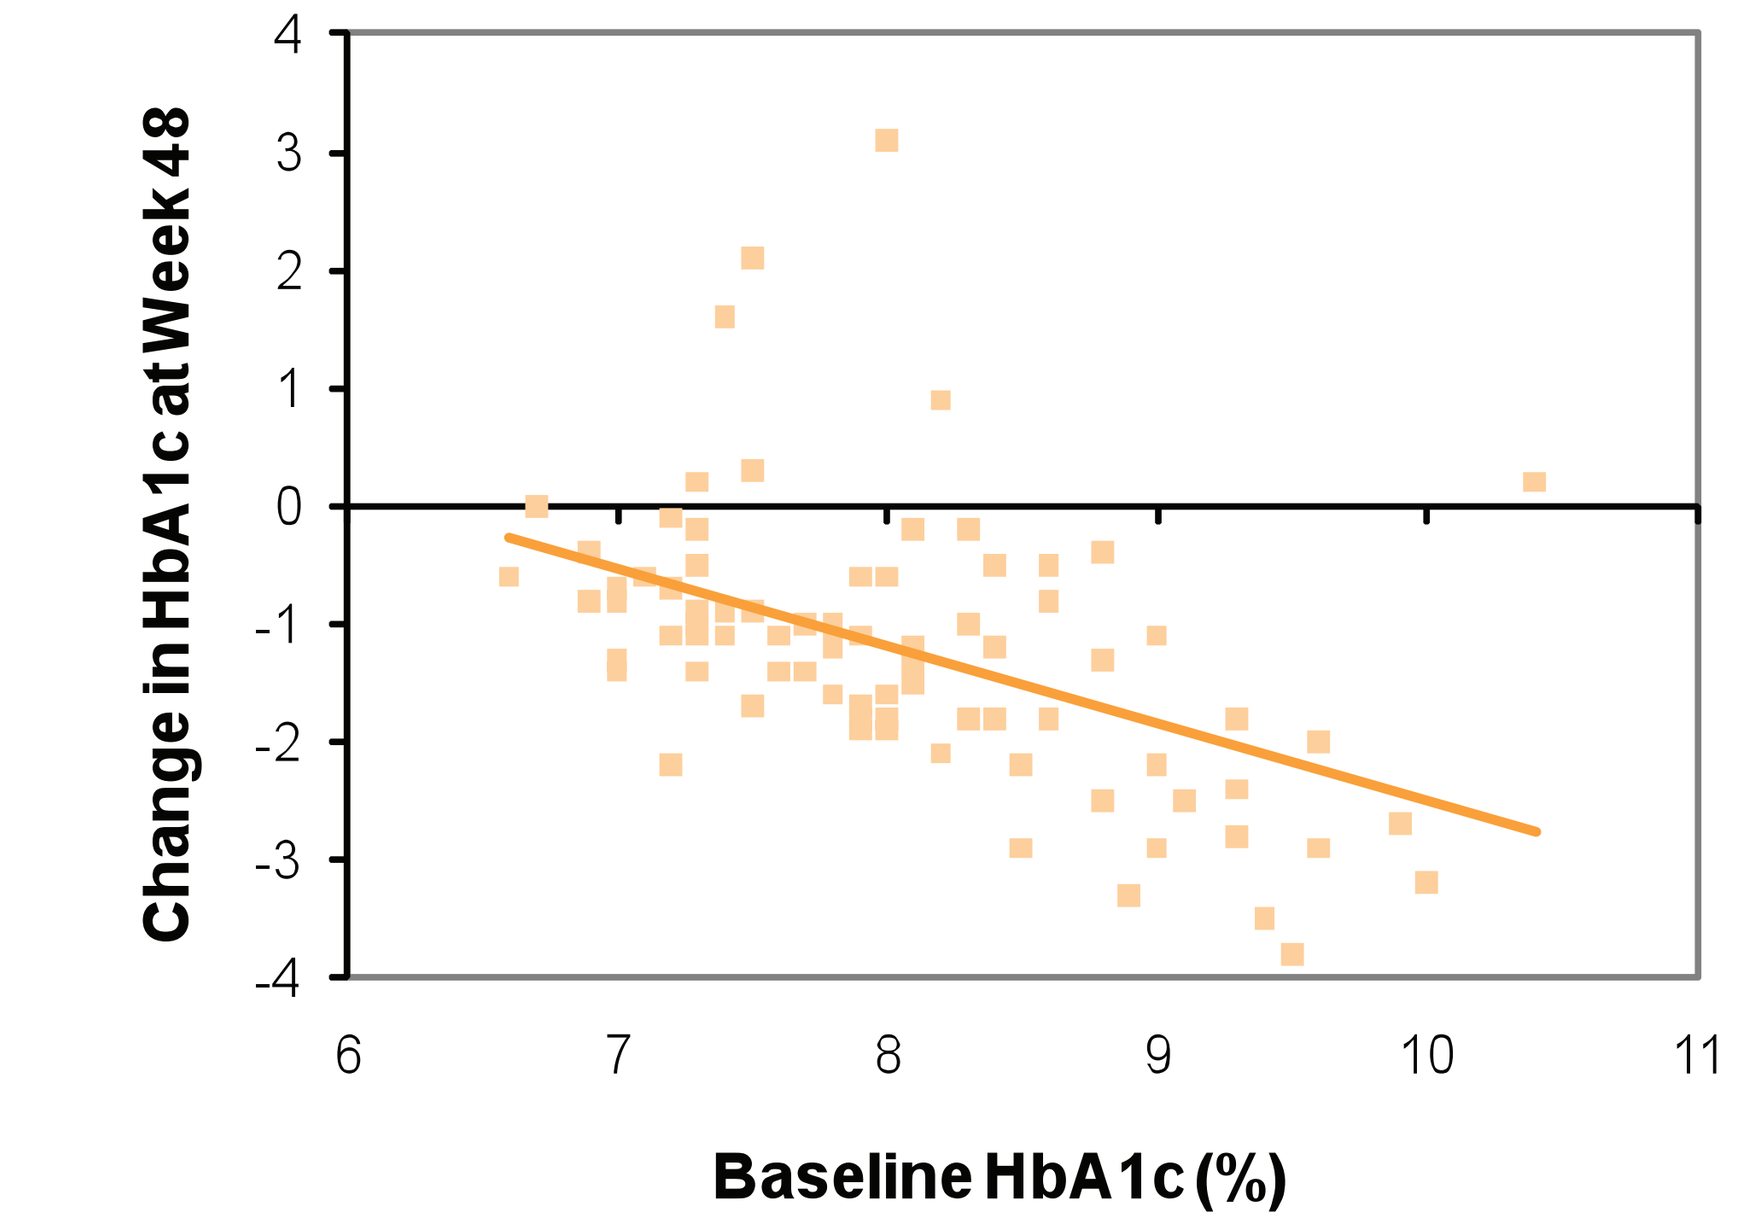
Change in HbA1c at Week 48
Baseline HbA1c (%)

Change in Body Weight Over Time

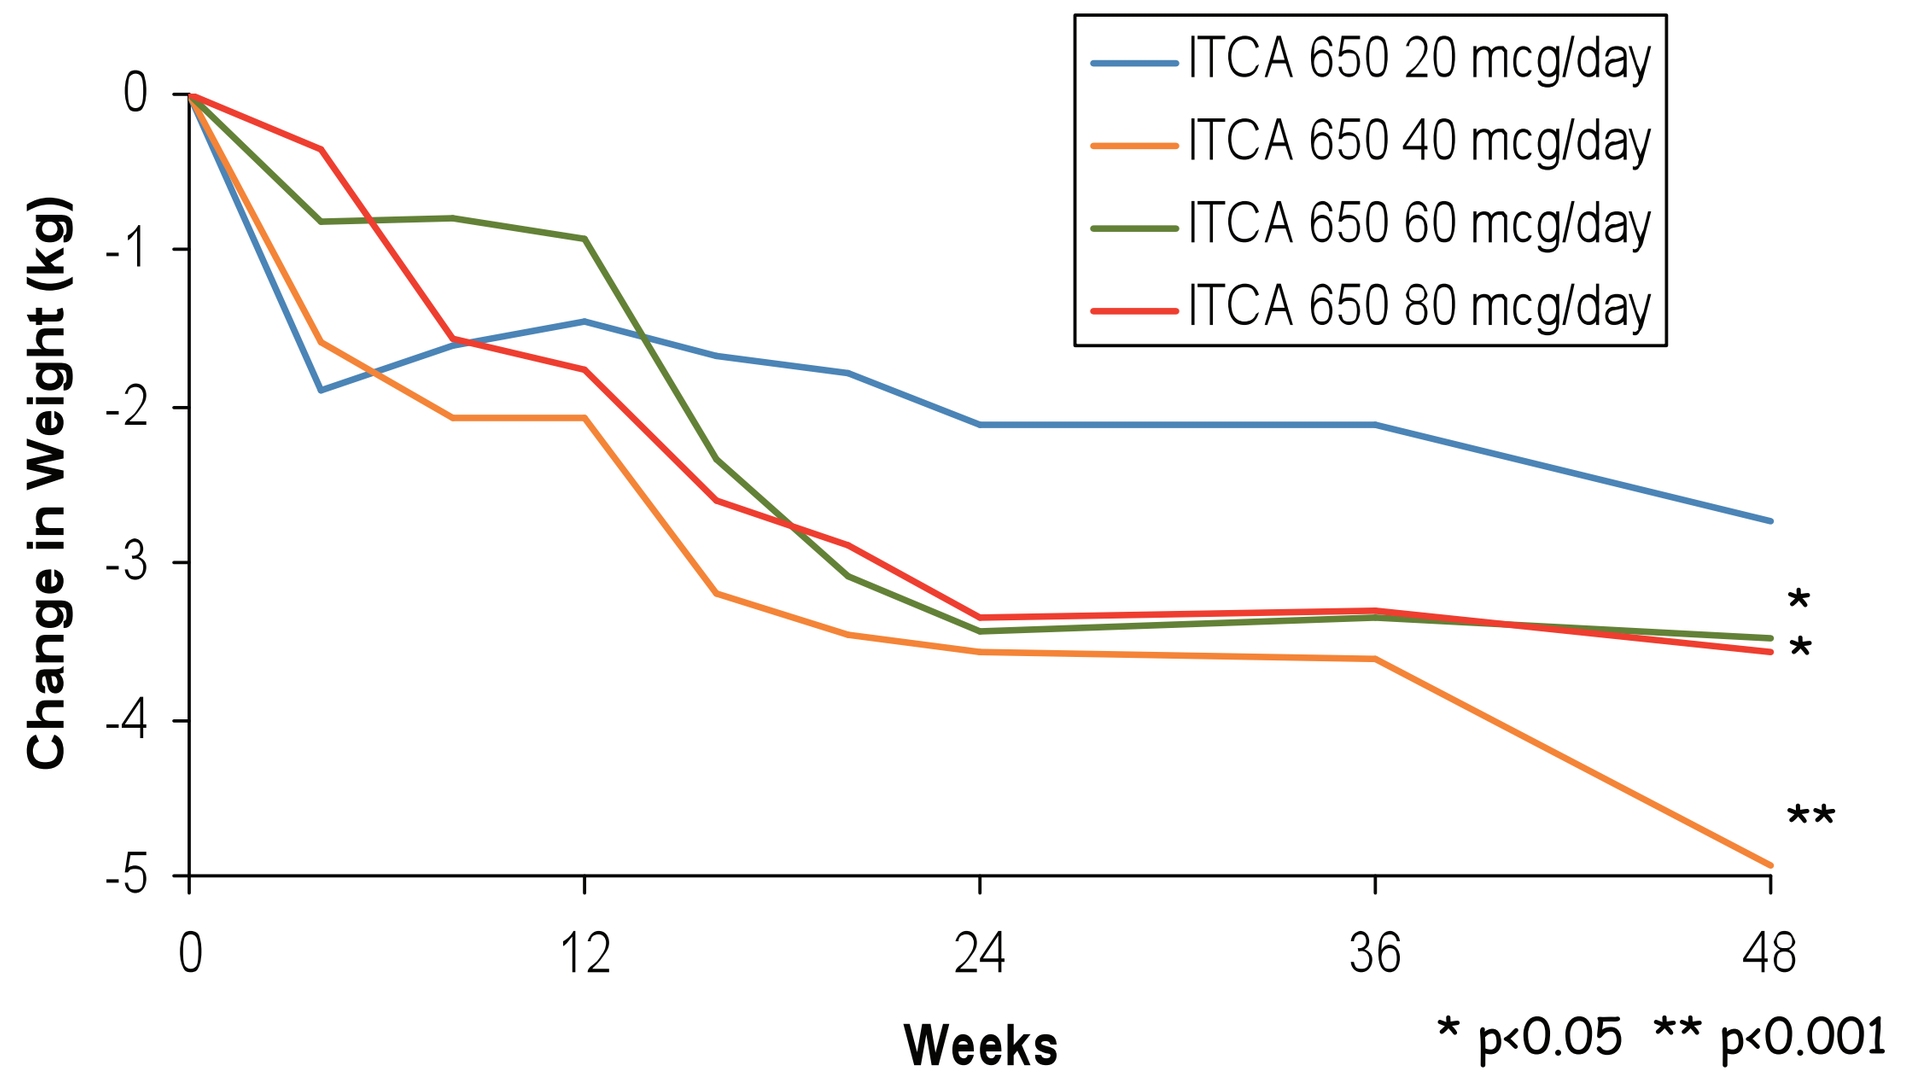
ITCA 650 20 mcg/day
ITCA 650 40 mcg/day
ITCA 650 60 mcg/day
ITCA 650 80 mcg/day
Change in Weight (kg)
Weeks
* p<0.05 ** p<0.001

Pharmacokinetics / Pharmacodynamics of ITCA 650

Day 1

- Plasma exenatide detected in 32 of 44 Phase 1 subjects within 6-12 hrs
- Steady state concentrations achieved within 24-48 hrs without an initial burst of drug
- Reductions in FPG observed within one day
- FPG and 2-hr postprandial glucose significantly decreased in the 20, 40 and 80 mcg/day dose groups within 1-5 days of initiating ITCA 650 and throughout the 28-day treatment period ($p<0.05$)

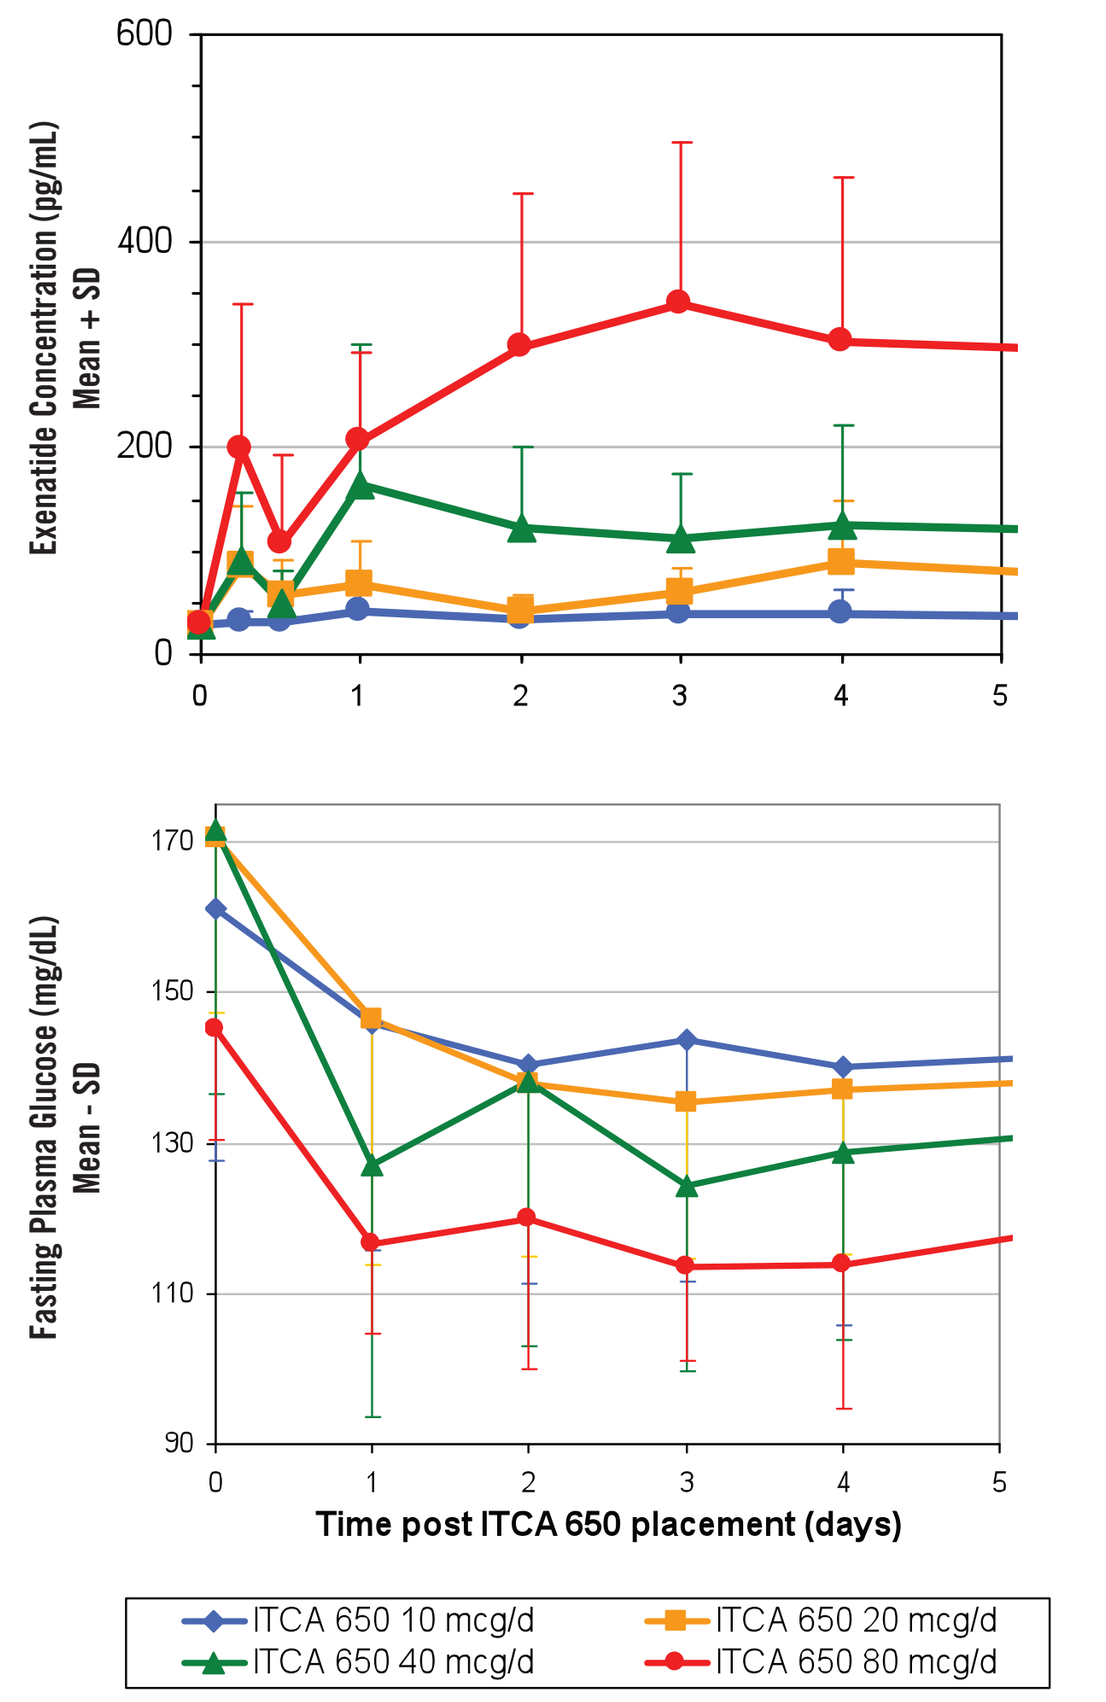
Exenatide Concentration (pg/mL) Mean + SD
Fasting Plasma Glucose (mg/dL) Mean - SD
Time post ITCA 650 placement (days)
ITCA 650 10 mcg/d
ITCA 650 20 mcg/d
ITCA 650 40 mcg/d
ITCA 650 80 mcg/d

Post Removal

- Consistent with its 2.4hr half-life, exenatide was not measurable in any subject 24 hrs after ITCA 650 removal
- Unlike other products with extended release delivery of exenatide, ITCA 650 allows immediate termination of delivery

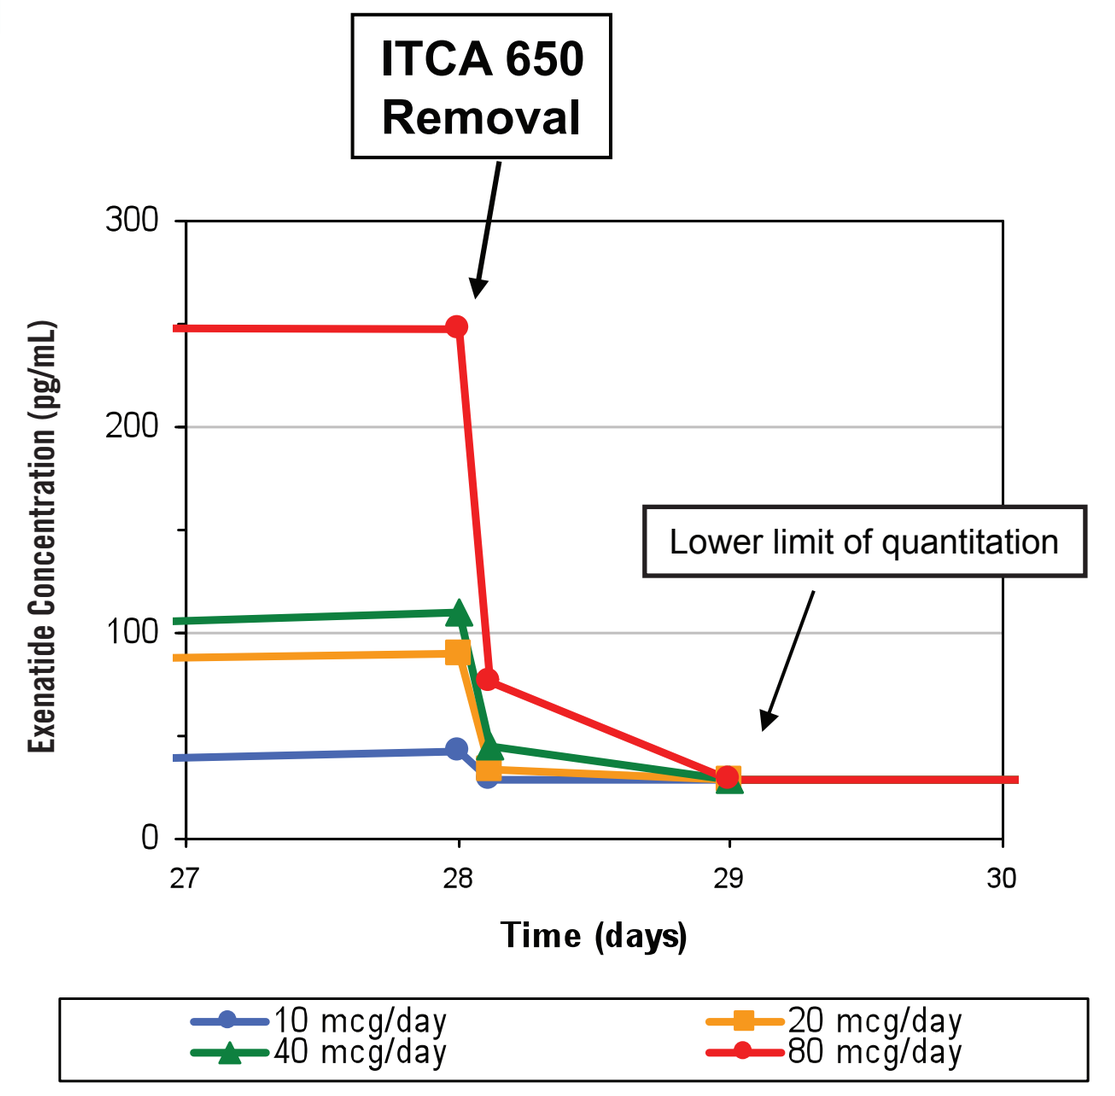
ITCA 650 Removal
Lower limit of quantitation
Exenatide Concentration (pg/mL)
Time (days)
10 mcg/day
40 mcg/day
20 mcg/day
80 mcg/day

PK Conclusions

- Dose-proportional exposure
- Reaches steady-state in 24-48 hours
- Following removal of ITCA 650, plasma exenatide levels drop rapidly to unquantifiable levels within 24 hours
- Provides continuous exposure with consistent plasma exenatide levels for up to 48 weeks

Weeks 1 through 12 / Weeks 13 through 48

Effect on HbA1c

- Significant decreases in HbA1c were observed in the first 12 weeks. Further decreases seen with longer time and higher concentrations.

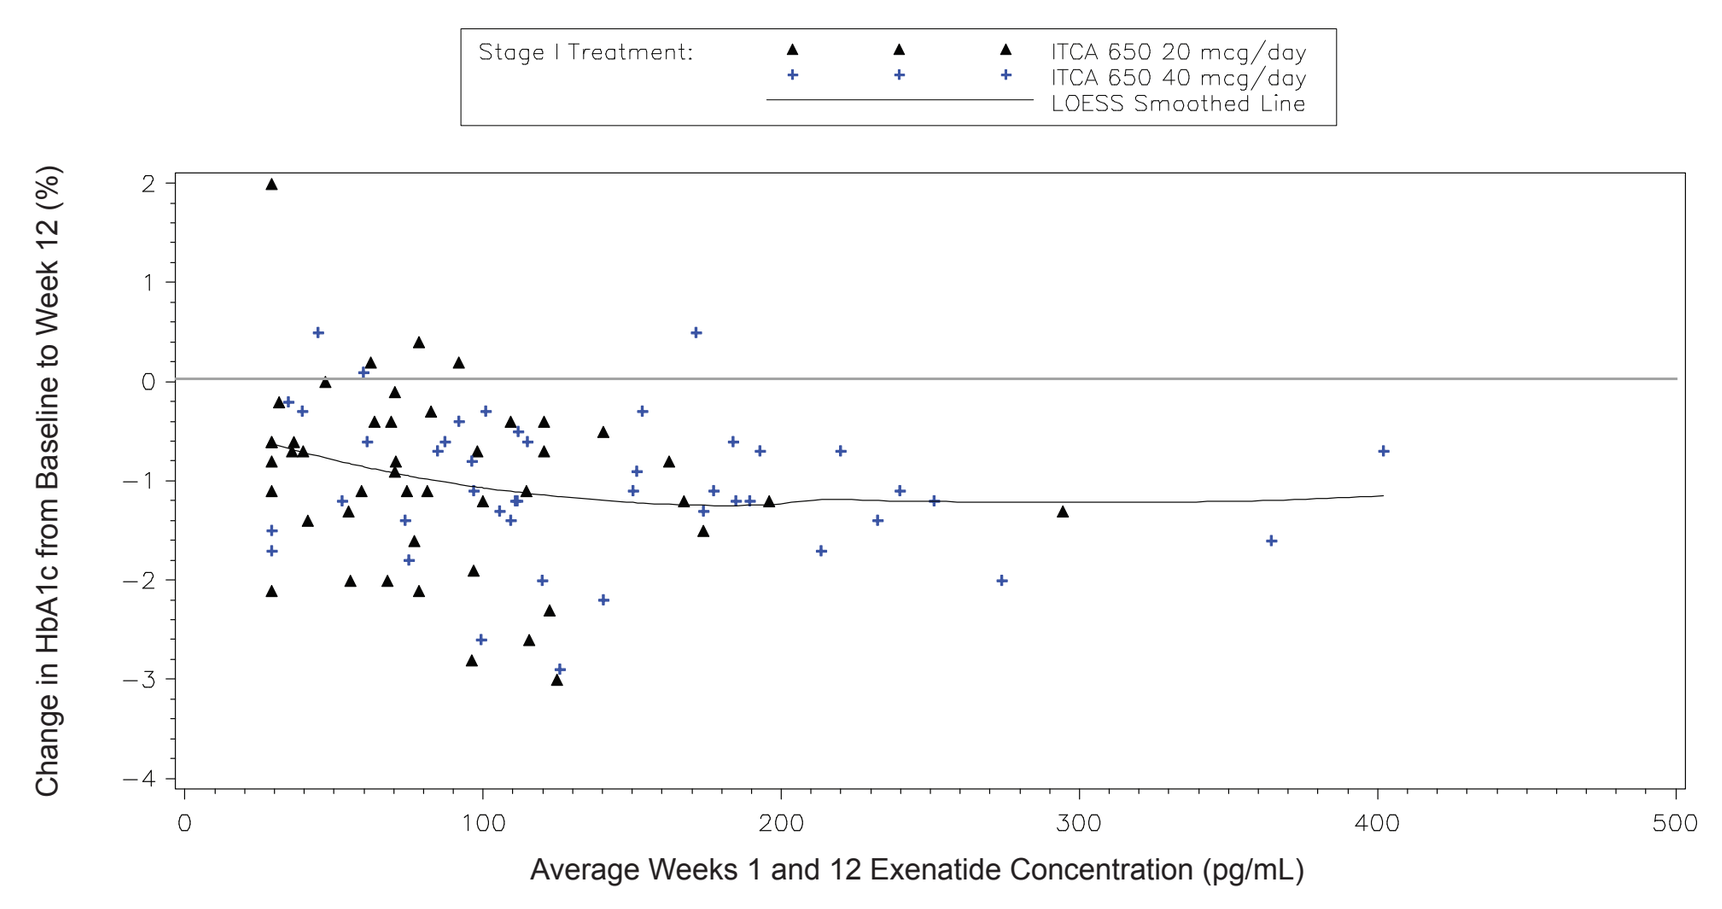
Stage I Treatment:
ITCA 650 20 mcg/day
ITCA 650 40 mcg/day
LOESS Smoothed Line
Change in HbA1c from Baseline to Week 12 (%)
Average Weeks 1 and 12 Exenatide Concentration (pg/mL)

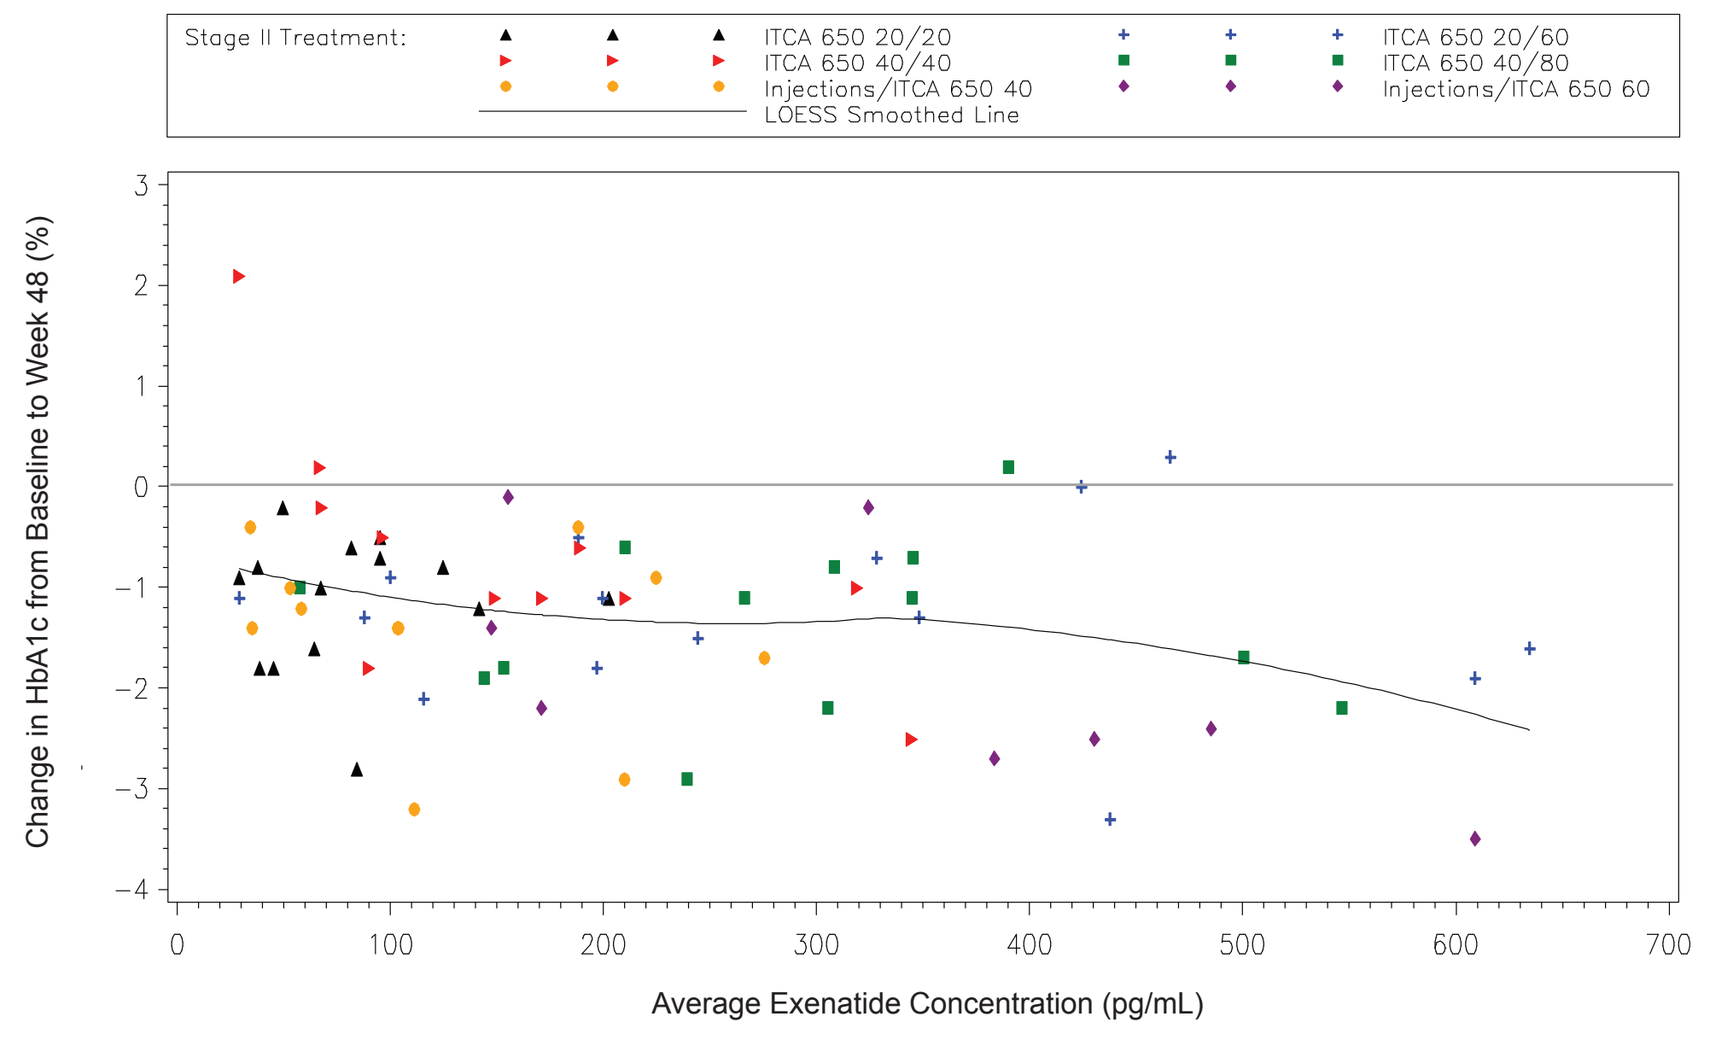
Stage II Treatment:
ITCA 650 20/20
ITCA 650 40/40
Injections/ITCA 650 40
LOESS Smoothed Line
ITCA 650 20/60
ITCA 650 40/80
Injections/ITCA 650 60
Change in HbA1c from Baseline to Week 48 (%)
Average Exenatide Concentration (pg/mL)

Effect on FPG

- Effects on FPG were seen in Weeks 1-12. Effect was maintained with longer time and higher doses.

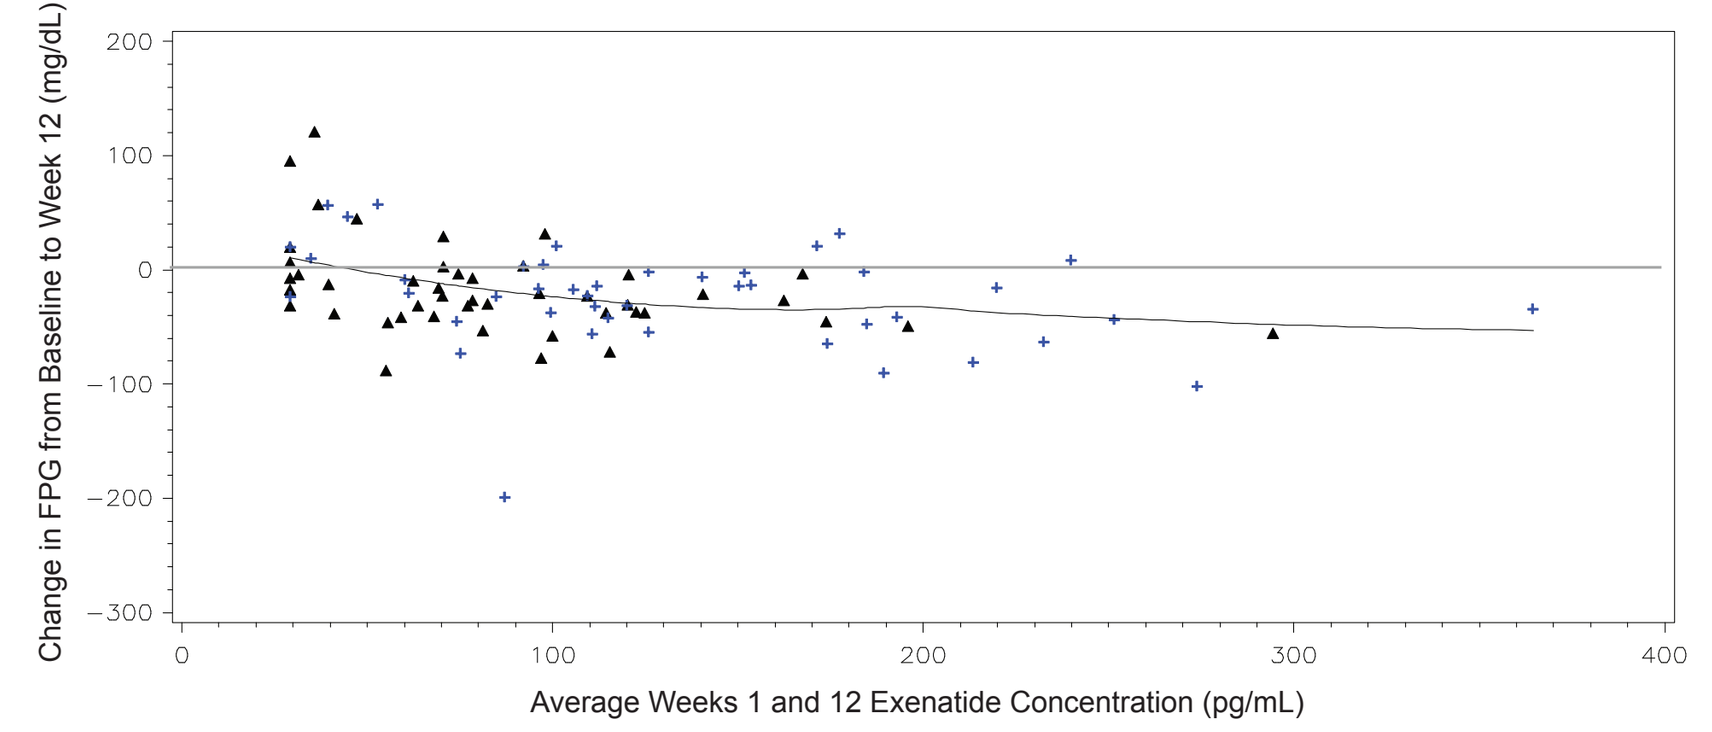
Change in FPG from Baseline to Week 12 (mg/dL)
Average Weeks 1 and 12 Exenatide Concentration (pg/mL)

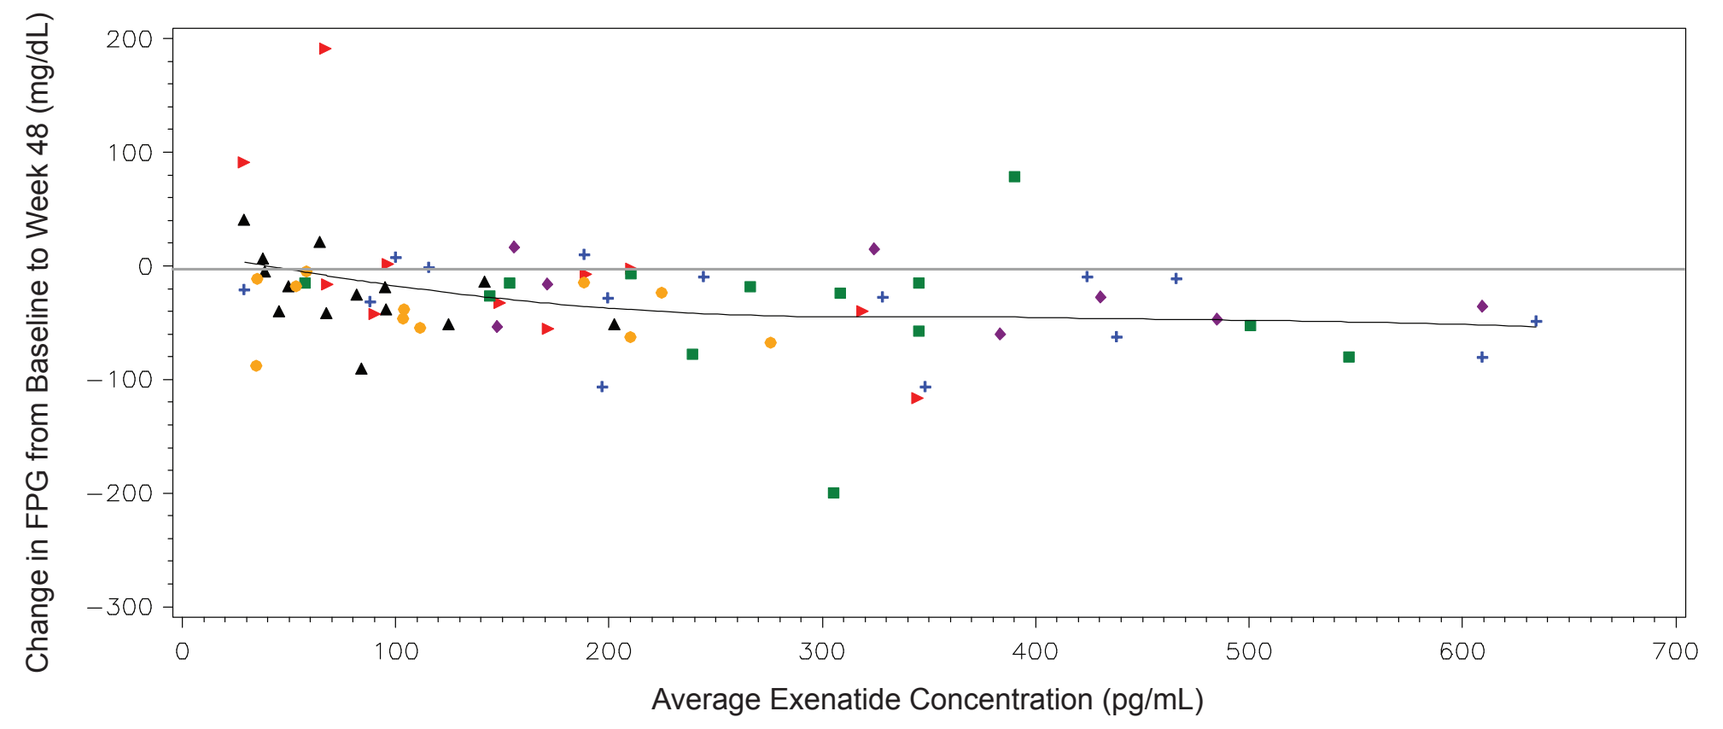
Change in FPG from Baseline to Week 48 (mg/dL)
Average Exenatide Concentration (pg/mL)

Effect on Weight

- Significant weight loss was observed in Weeks 1-12 with 86% of subjects experiencing weight loss by Week 24 (Week 24 data not shown). Some subjects lost >10% body weight.

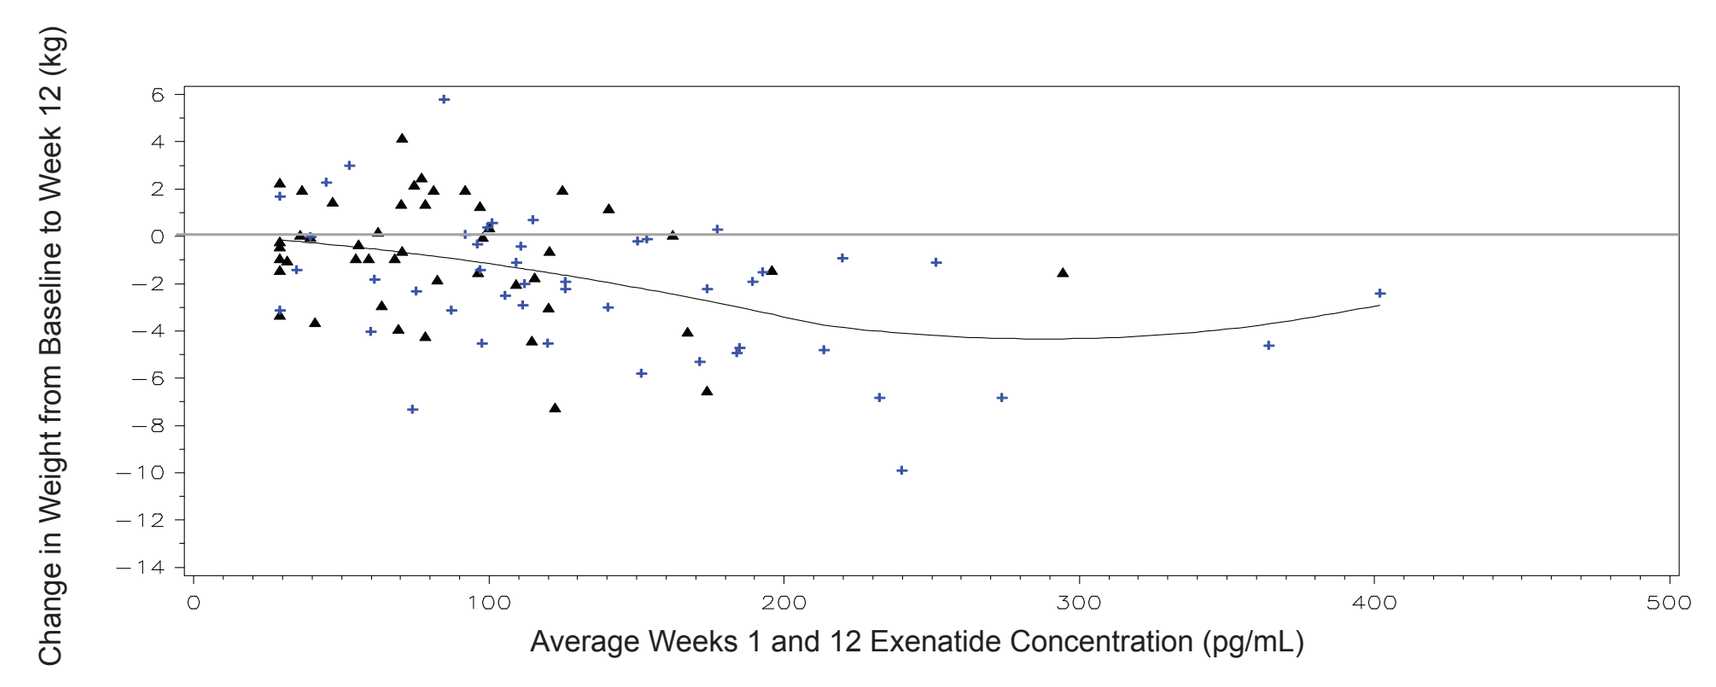
Change in Weight from Baseline to Week 12 (kg)
Average Weeks 1 and 12 Exenatide Concentration (pg/mL)

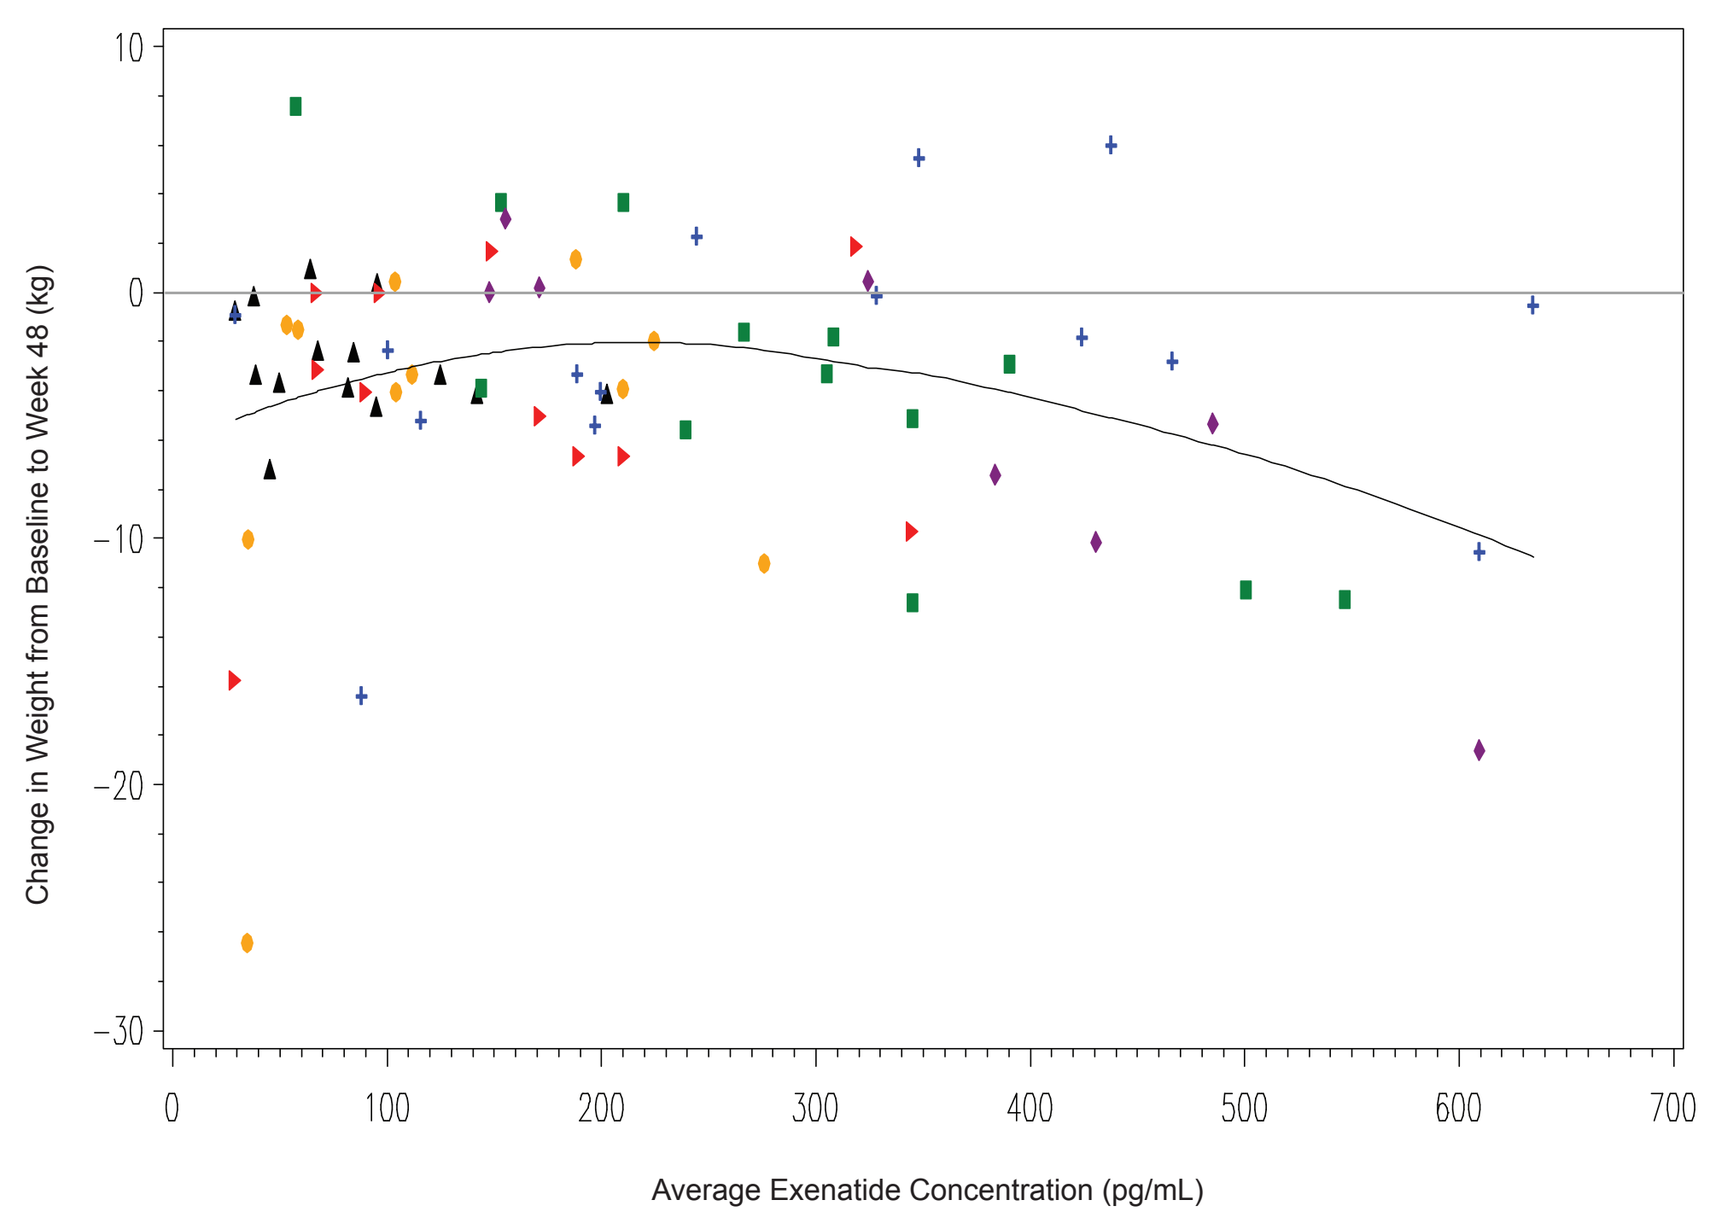
Change in Weight from Baseline to Week 48 (kg)
Average Exenatide Concentration (pg/mL)

PK/PD Conclusions

- All doses of ITCA 650 were active
- Higher exenatide concentrations tended to produce larger reductions in HbA1c, weight and FPG
 - Relatively flat concentration-response curve
 - Activity observed at exenatide levels < 30 pg/mL
- Nearly all patients responded to ITCA 650
 - Baseline HbA1c played a major role in determining magnitude of response to treatment
- Activity maintained throughout 48 weeks

Institutions:
1. Intarcia Therapeutics, Inc. Hayward, CA, USA
2. Biologistic Services, Boulder, CO, USA
3. Medpace, Cincinnati, OH, USA
4. University of California at San Diego, La Jolla, CA, USA
5. Dallas Diabetes and Endocrine Center, Dallas, TX, USA

Presented at the European Association for the Study of Diabetes September 12-16, 2011 in Lisbon, Portugal
This work was supported by Intarcia Therapeutics, Inc.
This poster will be made available online after the EASD meeting at http://www.intarcia.com/media

© 2011 Intarcia Therapeutics, Inc.